First long-term (two-year) controlled study to evaluate treatment with safinamide as add-on to levodopa in patients with Parkinson's disease and motor fluctuations

P05.287

Ravi Anand,[1] Rupam Borgohain,[2] Jozsef Attila Szasz,[3] Paolo Stanzione,[4] Chandrashekhar Meshram,[5] Mohit Bhatt,[6] Valentina Lucini[7]

[1]APC AG, St Moritz, Switzerland; [2]Nizam's Institute of Medical Sciences, Hyderabad, India; [3]Emergency County Hospital Targu Mures, Targu Mures, Romania; [4]Clinica Neurologica, Policlinico Tor Vergata, Rome, Italy; [5]Brain and Mind Institute, Nagpur, India; [6]Jaslok Hospital & Research Centre, Mumbai, India; [7]Newron Pharmaceuticals SpA, Bresso, Italy

INTRODUCTION

- Long-term use of levodopa in Parkinson's disease (PD) is limited by the development of motor fluctuations and dyskinesia,[1] estimated to affect approximately 40% of patients after 4–6 years of levodopa treatment.[2] Many of these patients require add-on therapy with another PD medication.
- Safinamide is an α-aminoamide in development as add-on therapy to dopamine agonists in early PD[3,4] and to levodopa in mid- to late-stage PD.[5] Safinamide has both dopaminergic and non-dopaminergic mechanisms of action, including selective inhibition of monoamine oxidase-B (MAO-B), blockade of Na^+ channels, and inhibition of glutamate release *in vitro*.[6-9]
- Six months' treatment with safinamide (50 and 100 mg/day) as add-on to levodopa in patients with PD and motor fluctuations significantly improved ON time with no/minor dyskinesia and motor function versus placebo (Study 016).

OBJECTIVE

- Study 018 was an 18-month, placebo-controlled extension to Study 016. Its aim was to assess the long-term efficacy and safety of safinamide as add-on to levodopa in patients with PD and motor fluctuations.

METHODS

Study design

- Study 018 was a double-blind, placebo-controlled, 18-month extension to the pivotal Phase III Study 016 (Figure 1).

Figure 1. Study design

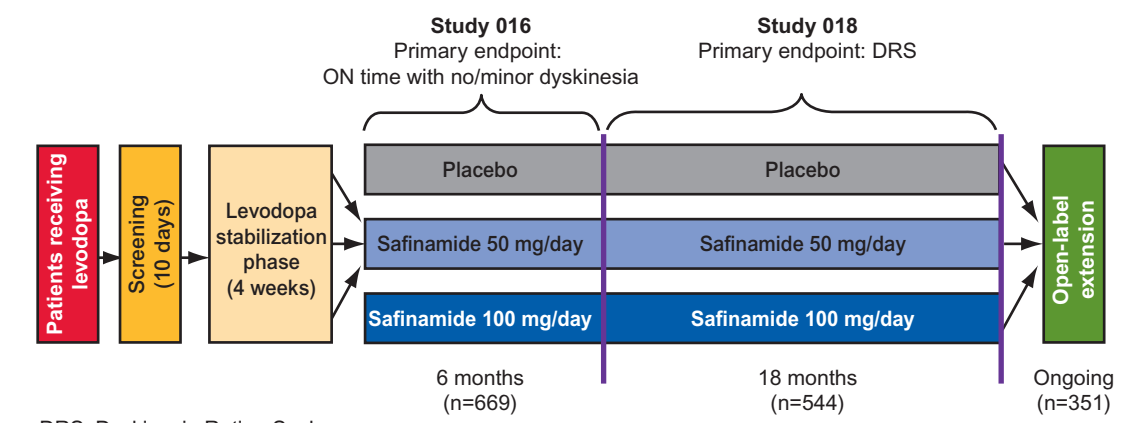
Study 016
Primary endpoint:
ON time with no/minor dyskinesia
Study 018
Primary endpoint: DRS
Patients receiving levodopa
Screening (10 days)
Levodopa stabilization phase (4 weeks)
Placebo
Placebo
Safinamide 50 mg/day
Safinamide 50 mg/day
Safinamide 100 mg/day
Safinamide 100 mg/day
Open-label extension
6 months (n=669)
18 months (n=544)
Ongoing (n=351)

DRS, Dyskinesia Rating Scale

Patients

- At recruitment into Study 016, patients were: male or female, aged 30−80 years, with a 3-year history of idiopathic PD, a Hoehn and Yahr staging of I–IV during an OFF phase, receiving treatment with a stable dose of levodopa, and experiencing motor fluctuations with >1.5 hours of OFF time during the day.
- To be eligible for inclusion in Study 018, patients were required to have completed 24 weeks' treatment in Study 016 and to be compliant with study medication. Patients with clinically significant adverse events (AEs) or Hoehn and Yahr Stage V were not eligible.
- Patients continued to take the same dose of safinamide that they took at the end of Study 016. Patients who were unable to tolerate safinamide 100 mg/day had their dose decreased to 50 mg/day.
- The dose of levodopa and any other PD therapies could be changed during the 18-month period and other PD medications (excluding MAO inhibitors) could be added, if necessary. Patients who had their dose increased by ⩾20% (or a new PD drug added) had their data censored at this time ('on treatment' analysis).

Efficacy measures

- The primary endpoint was change from baseline (start of Study 016) to Month 24 in Dyskinesia Rating Scale (DRS) scores during ON time.
- Some secondary and tertiary endpoints included:
 - ON time with no/minor dyskinesia
 - Unified Parkinson's Disease Rating Scale (UPDRS) Part IV (complications of therapy) total score
 - UPDRS Part II (activities of daily living [ADL]) scores and response rates
 - UPDRS Part III (motor) scores
 - Clinical Global Impression-Change (CGI-C) and -Severity (CGI-S)
 - Change in individual diary categories (ON with no dyskinesia, ON with minor dyskinesia, ON with troublesome dyskinesia, OFF, asleep)
 - GRID-Hamilton Depression Rating Scale (GRID-HAMD)
 - 39-item Parkinson's Disease Questionnaire (PDQ-39) subscale scores.

Post-hoc analysis

- A post-hoc analysis was performed in the subgroup of patients with DRS scores >4 at baseline.

Safety measures

- Safety and tolerability were assessed using AEs, and laboratory, vital sign, ophthalmologic, and electrocardiogram (ECG) data.

Statistical analysis

- Analysis of efficacy measures was hierarchical; if the primary endpoint was not significant, subsequent endpoints were considered exploratory.
- The primary endpoint was analyzed using a mixed linear model.
- Mean change in ADL (UPDRS Part II), change in diary times, change in UPDRS Parts III and IV, and CGI-S were analyzed using an analysis of covariance (ANCOVA).
- Post-hoc analysis of DRS scores in patients with baseline DRS >4 was performed using ANCOVA.
- The intention-to-treat (ITT) population was defined as all patients who were randomized into Study 016. The ITT population, during 'on treatment', was the primary analysis population for efficacy.
- The safety population was defined as all patients who received at least one dose of study medication and who had at least one post-dose safety assessment.

RESULTS

Patients

- 544 of the 669 patients in Study 016 entered Study 018: 175/222 (78.8%) placebo, 189/223 (84.8%) safinamide 50 mg/day, and 180/224 (80.4%) safinamide 100 mg/day.
- 440 of the 544 patients completed 24 months of treatment: 142/175 (81.1%) placebo, 148/189 (78.3%) safinamide 50 mg/day, and 150/180 (83.8%) safinamide 100 mg/day.
- Patient baseline characteristics are shown in Table 1.

Table 1. Baseline patient demographics and clinical characteristics

Characteristic	Placebo (N=222)	Safinamide 50 mg/day (N=223)	Safinamide 100 mg/day (N=224)
Gender, male, n (%)	160 (72.1)	157 (70.4)	163 (72.8)
Race, n (%)			
Asian	180 (81.1)	180 (80.7)	179 (79.9)
White	42 (18.9)	43 (19.3)	45 (20.1)
Age, years, mean (SD)	59.4 (9.41)	60.1 (9.65)	60.1 (9.19)
H&Y Stage, mean (SD)	2.8 (0.7)	2.8 (0.6)	2.8 (0.6)
Disease duration, years, mean (SD)	8.3 (3.8)	7.9 (4.0)	8.2 (3.8)
Daily OFF time, hours, mean (SD)	5.3 (2.1)	5.2 (2.1)	5.2 (2.2)
Daily ON time, hours, mean (SD)	9.3 (2.2)	9.4 (2.3)	9.5 (2.4)
UPDRS III score ON, mean (SD)	28.7 (12.0)	27.3 (12.7)	28.3 (13.3)
Concomitant PD medication, n (%)			
Levodopa	222 (100)	223 (100)	224 (100)
Dopamine agonist	137 (61.7)	142 (63.7)	128 (57.1)
Entacapone	56 (25.2)	52 (23.3)	55 (24.6)
Anticholinergic	87 (39.2)	74 (33.2)	87 (38.8)
Amantadine	34 (15.3)	29 (13.0)	30 (13.4)
Baseline DRS scores, mean (SD)			
ITT population	3.4 (3.93)	3.9 (3.89)	3.7 (4.07)
DRS >4[a]	7.9 (3.08)	8.0 (2.74)	8.4 (2.81)

[a]Placebo (N=76), safinamide 50 mg/day (N=86), safinamide 100 mg (N=80)
DRS, Dyskinesia Rating Scale; H&Y, Hoehn and Yahr; ITT, intention to treat; PD, Parkinson's disease; SD, standard deviation; UPDRS, Unified Parkinson's Disease Rating Scale

Efficacy

- In the overall study population, there was a worsening in DRS score for placebo and an improvement in the safinamide groups but this did not reach statistical significance (Figure 2a; p=0.21 and 0.15 for safinamide 50 and 100 mg/day, respectively).
- Safinamide 50 and 100 mg/day increased the average daily ON time with no/minor dyskinesia by 0.67 and 0.83 hours, respectively, compared with placebo at Month 24 (Figure 3).
- Total mean daily OFF time was reduced by 0.62 and 0.75 hours, respectively, in the safinamide 50 and 100 mg/day groups, compared with placebo at Month 24.

Figure 2. DRS scores during ON in the a) ITT population (primary endpoint) and b) baseline DRS >4 (post hoc)

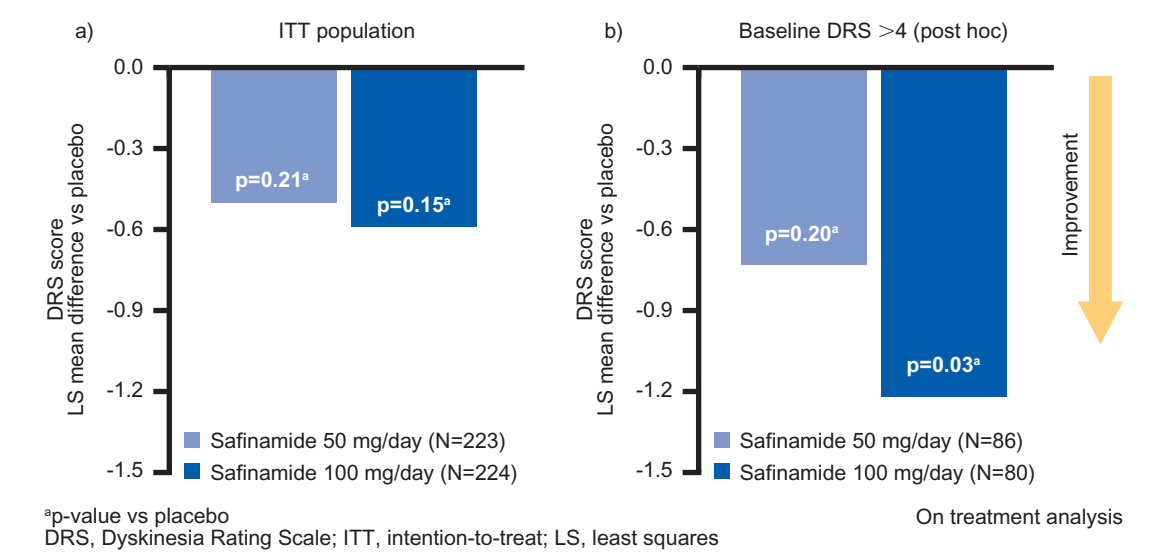
a)
ITT population
b)
Baseline DRS >4 (post hoc)
LS mean difference vs placebo DRS score
0.0
-0.3
-0.6
-0.9
-1.2
-1.5
p=0.21[a]
p=0.15[a]
p=0.20[a]
p=0.03[a]
Improvement
Safinamide 50 mg/day (N=223)
Safinamide 100 mg/day (N=224)
Safinamide 50 mg/day (N=86)
Safinamide 100 mg/day (N=80)
On treatment analysis

[a]p-value vs placebo
DRS, Dyskinesia Rating Scale; ITT, intention-to-treat; LS, least squares

Figure 3. ON time with no/minor dyskinesia (ITT population)

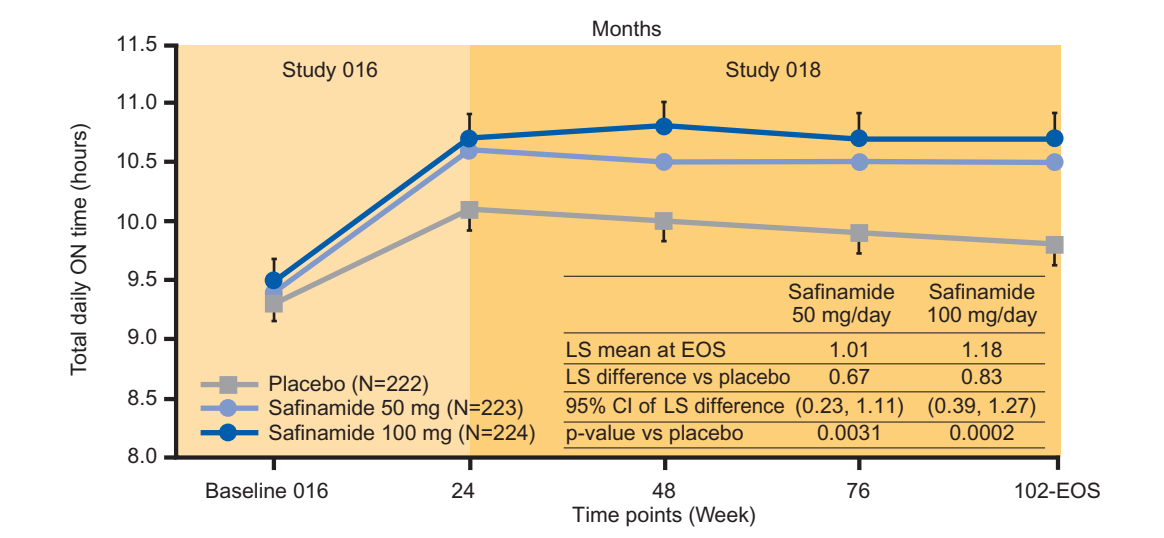
Months
Study 016
Study 018
Total daily ON time (hours)
11.5
11.0
10.5
10.0
9.5
9.0
8.5
8.0
Baseline 016
24
48
76
102-EOS
Time points (Week)
Placebo (N=222)
Safinamide 50 mg (N=223)
Safinamide 100 mg (N=224)
Safinamide 50 mg/day | Safinamide 100 mg/day
LS mean at EOS 1.01 1.18
LS difference vs placebo 0.67 0.83
95% CI of LS difference (0.23, 1.11) (0.39, 1.27)
p-value vs placebo 0.0031 0.0002

CI, confidence interval; EOS, end of study; LS, least squares

- At Month 24, scores for UPDRS Part II and III, CGI-S, GRID-HAMD, and several PDQ-39 subscale scores showed improvements for safinamide 100 mg/day versus placebo (Table 2).

Table 2. Key secondary and tertiary endpoints

	24 weeks (Study 016) (N=669)		24 months (Study 018) (N=669)	
	Safinamide 50 mg/day	Safinamide 100 mg/day	Safinamide 50 mg/day	Safinamide 100 mg/day
LS mean change vs placebo				
ON time	0.51*	0.55*	0.67**	0.83***
OFF time	-0.6**	-0.6**	-0.62**	-0.75***
UPDRS II	-0.5	-1.0**	-0.52	-1.06**
UPDRS III	-1.8*	-2.6***	-1.05	-2.13**
UPDRS IV	-0.3(*)	-0.6***	-0.12	-0.68***
CGI-Severity	-0.2**	-0.1*	-0.16**	-0.14*
GRID-HAMD	-0.2	-0.5(*)	-0.15	-0.57*
PDQ-39				
Mobility	-2.4	-2.0	-2.5	-2.5(*)
ADL	-3.5*	-2.6(*)	-4.9**	-3.5*
Emotional wellbeing	-0.7	-3.4*	-1.8	-3.8**
Stigma	-1.4	-0.4	-1.4	-0.4
Social support	1.6	-0.1	0.5	-1.1
Cognition	1.3	-1.1	-0.4	-0.6
Communication	-1.5	-3.3*	-2.8(*)	-3.0*
Bodily discomfort	1.1	-3.7*	1.9	-3.7*
% patients with improvement				
CGI-Change[a]	66.4%***	64.3%**	62.3%**	59.8%(*)

(*)p<0.1; *p<0.05; **p⩽0.01; ***p<0.001;[a]% in placebo group were 55.4% (Week 24) and 53.6% (Month 24)
CGI, Clinical Global Impression; GRID-HAMD, GRID-Hamilton Depression Rating Scale; LS, least squares; PDQ-39, 39-item Parkinson's Disease Questionnaire; UPDRS, Unified Parkinson's Disease Rating Scale

Post-hoc analysis

- At Month 24, least squares mean changes in DRS scores compared with placebo (N=76) were -0.73 (50 mg/day; p=0.1999; N=86) and -1.22 (100 mg/day; p=0.0317; N=80) in the subgroup of patients who had DRS scores >4 at baseline (Figure 2b; 'on treatment').
- The number (%) of patients with baseline DRS >4 who had a reduction in levodopa dose during the study was 12 (15.8%), 17 (19.8%), and 21 (26.3%) in the placebo, safinamide 50 mg/day, and safinamide 100 mg/day groups, respectively (p=0.0585 for 100 mg/day vs placebo).

Safety

- The most frequent treatment-emergent AEs for Studies 016 and 018 combined (for all 669 patients) are reported in Table 3a. The most common newly emergent AEs observed during Study 018 (for the 544 patients) are reported in Table 3b.

Table 3. a) Most frequent treatment-emergent adverse events for Studies 016 and 018 combined; b) newly emergent[a] adverse events (>5% in any group) during Study 018

a)

Adverse event (preferred term)	Placebo (N=175)			Safinamide 50 mg/day (N=189)			Safinamide 100 mg/day (N=180)		
	n	%	% severe	n	%	% severe	n	%	% severe
Dyskinesia	38	21.7	2.9	59	31.2	2.1	50	27.8	1.7
Parkinson's disease	42	24.0	1.1	42	22.2	0.5	43	23.9	0.0
Cataract	27	15.4	0.0	27	14.3	0.5	25	13.9	0.0
Back pain	21	12.0	0.0	17	9.0	0.5	23	12.8	1.1
Asthenia	21	12.0	0.0	14	7.4	0.0	21	11.7	0.0
Pyrexia	21	12.0	1.1	22	11.6	0.0	15	8.3	0.0
Insomnia	11	6.3	0.6	21	11.1	0.0	13	7.2	0.0
Headache	13	7.4	0.0	20	10.6	0.0	15	8.3	0.0
Fall	17	9.7	1.7	20	10.6	1.1	15	8.3	1.7
Hypertension	12	6.9	0.0	19	10.1	0.0	18	10.0	0.0

b)

Adverse event	Placebo (N=175) n (%)	Safinamide 50 mg/day (N=189) n (%)	Safinamide 100 mg/day (N=180) n (%)
Parkinson's disease	29 (16.6)	32 (16.9)	38 (21.1)
Dyskinesia	27 (15.4)	24 (12.7)	24 (13.3)
Cataract	18 (10.3)	21 (11.1)	18 (10.0)
Asthenia	13 (7.4)	10 (5.3)	16 (8.9)
Pyrexia	13 (7.4)	14 (7.4)	11 (6.1)
Fall	13 (7.4)	13 (6.9)	11 (6.1)
Back pain	9 (5.1)	9 (4.8)	14 (7.8)
Weight decreased	8 (4.6)	13 (6.9)	8 (4.4)
Constipation	8 (4.6)	12 (6.3)	9 (5.0)
Insomnia	7 (4.0)	18 (9.5)	6 (3.3)
Hypertension	6 (3.4)	7 (3.7)	11 (6.1)
Pain intensity	5 (2.9)	10 (5.3)	7 (3.9)
Arthralgia	5 (2.9)	10 (5.3)	4 (2.2)

[a]Any adverse event (AE) not reported in Study 016 or an AE reported in Study 016 that occurred with greater intensity in Study 018

- There were no clinically relevant differences in vital signs, laboratory tests, ophthalmologic assessments, or ECG recordings between groups (approximately 50% of patients in each group presented at least one laboratory/clinically notable value) during Study 018.
- Discontinuations due to AEs, serious AEs, deaths, and discontinuations due to AEs over two years were similar across all groups (Table 4).

Table 4. Notable events over two years of treatment

Event, n (%)	Placebo (N=222)	Safinamide 50 mg/day (N=223)	Safinamide 100 mg/day (N=224)
Discontinuations due to AEs	23 (10.4)	20 (9.0)	30 (13.4)
Serious AEs	37 (16.7)	35 (15.7)	45 (20.1)
Deaths	8 (3.6)	5 (2.2)	12 (5.4)

AE, adverse event

CONCLUSION

- **This is the first two-year, prospective, placebo-controlled study of the efficacy and safety of safinamide in patients with mid-late PD and motor fluctuations, despite optimized antiparkinsonian treatment.**
- **At Month 24, there was no significant difference in DRS scores (p=0.21 and 0.15 for safinamide 50 and 100 mg/day, respectively), while the other efficacy measures improved. This may have been due to the low baseline DRS scores and the low incidence of troublesome dyskinesia at baseline. In the post-hoc analysis of the subgroup of patients with DRS >4, safinamide 100 mg/day improved DRS scores versus placebo (p=0.03).**
- **Safinamide 50 and 100 mg/day improved ON time with no/minor dyskinesia and reduced total daily OFF time when used as add-on to levodopa in patients with PD and motor fluctuations, despite optimized PD therapy, as shown by patient diary data; the improvements seen at 6 months were still present at 2 years. In the safinamide 100 mg/day group, these were accompanied by long-term improvements in motor function (UPDRS Part III), non-motor symptoms (ADL, depressive symptoms), and patients' clinical status.**
- **Completion rates were high and no new safety concerns emerged with either dose of safinamide in this extension study.**

REFERENCES

1. Hauser RA. Eur Neurol 2009; 62: 1-8.
2. Ahlskog JE and Muenter MD. Mov Disord 2001; 16: 448-458.
3. Stocchi F et al. Neurology 2004; 63: 746-748.
4. Stocchi F et al. Neurology 2007; 68 (12 Suppl 1): A109.
5. Borgohain R et al. Parkinsonism Relat Disord 2009; 15 (Suppl 2): S115.
6. Pevarello P et al. J Med Chem 1998; 41: 579-590.
7. Caccia C et al. Neurology 2006; 67 (7 Suppl 2): S18-S23.
8. Caccia C et al. Parkinsonism Relat Disord 2007; 13 (Suppl 2): S99.
9. Chazot PL. Curr Opin Investig Drugs 2007; 8: 570-579.

This study was funded by Newron and Merck Serono S.A. – Geneva, an affiliate of Merck KGaA Darmstadt, Germany.

The authors thank Jon Stamford of Complete Medical Communications (supported by Merck Serono S.A. – Geneva, Switzerland, an affiliate of Merck KGaA, Darmstadt, Germany) for writing assistance.

The Study 018 investigators were:

ITALY: T.P. Avarello, U. Bonuccelli, G. Fabbrini, R. Marconi, L. Morgante, M. Onofrj, C. Pacchetti, P. Stanzione, F. Stocchi.

ROMANIA: O. Bajenaru, A. Bulboaca, A. Campeanu, D. Chirileanu, D. Muresanu, C. Panea, C. Popescu, M. Simu, J. Szasz, M. Ticmeanu.

INDIA: S. Bandishti, R. Bansal, M. Behari, M. Bhatt, R. Borgohain, P. Chakraborty, Dwivedee, N. Ichoporia, Karadan, Keshwa, A. Kishore, S. Kothari, S. Kuman, A. Kumar, A. Kumar Roy, Mehndiratta, N. Mehta, C. Meshram, U.K. Misra, J.M.K Murthy, A. Pangaria, Prabhakar, Pradhan, P. Pal, V.S. Prasad, C. Prasad Das, Sahadulla, C. Saukhla, P.K. Sethi, A. Shah, Shankara, R. Shukla, Y. Singh, A. Sowani, R. Srinivasa, A.V. Srinivasan, C.J. Vaas, Vasudevan, Velmurgendran, Verma, Vijayan

American Academy of Neurology Annual Meeting, Honolulu, Hawaii, USA, 9-16 April 2011